Case-matched comparison of intramuscular versus vaginal progesterone for luteal phase support after in vitro fertilization and embryo transfer

Naveed Khan, M.D.,[a] Kevin S. Richter, Ph.D.,[a] Emily J. Blake, M.D.,[b] Vladimir I. Yankov, M.D.[b]

[a] Shady Grove Fertility Reproductive Science Center, Rockville, MD, and [b] Ferring Pharmaceuticals, Inc., Suffern, NY, USA

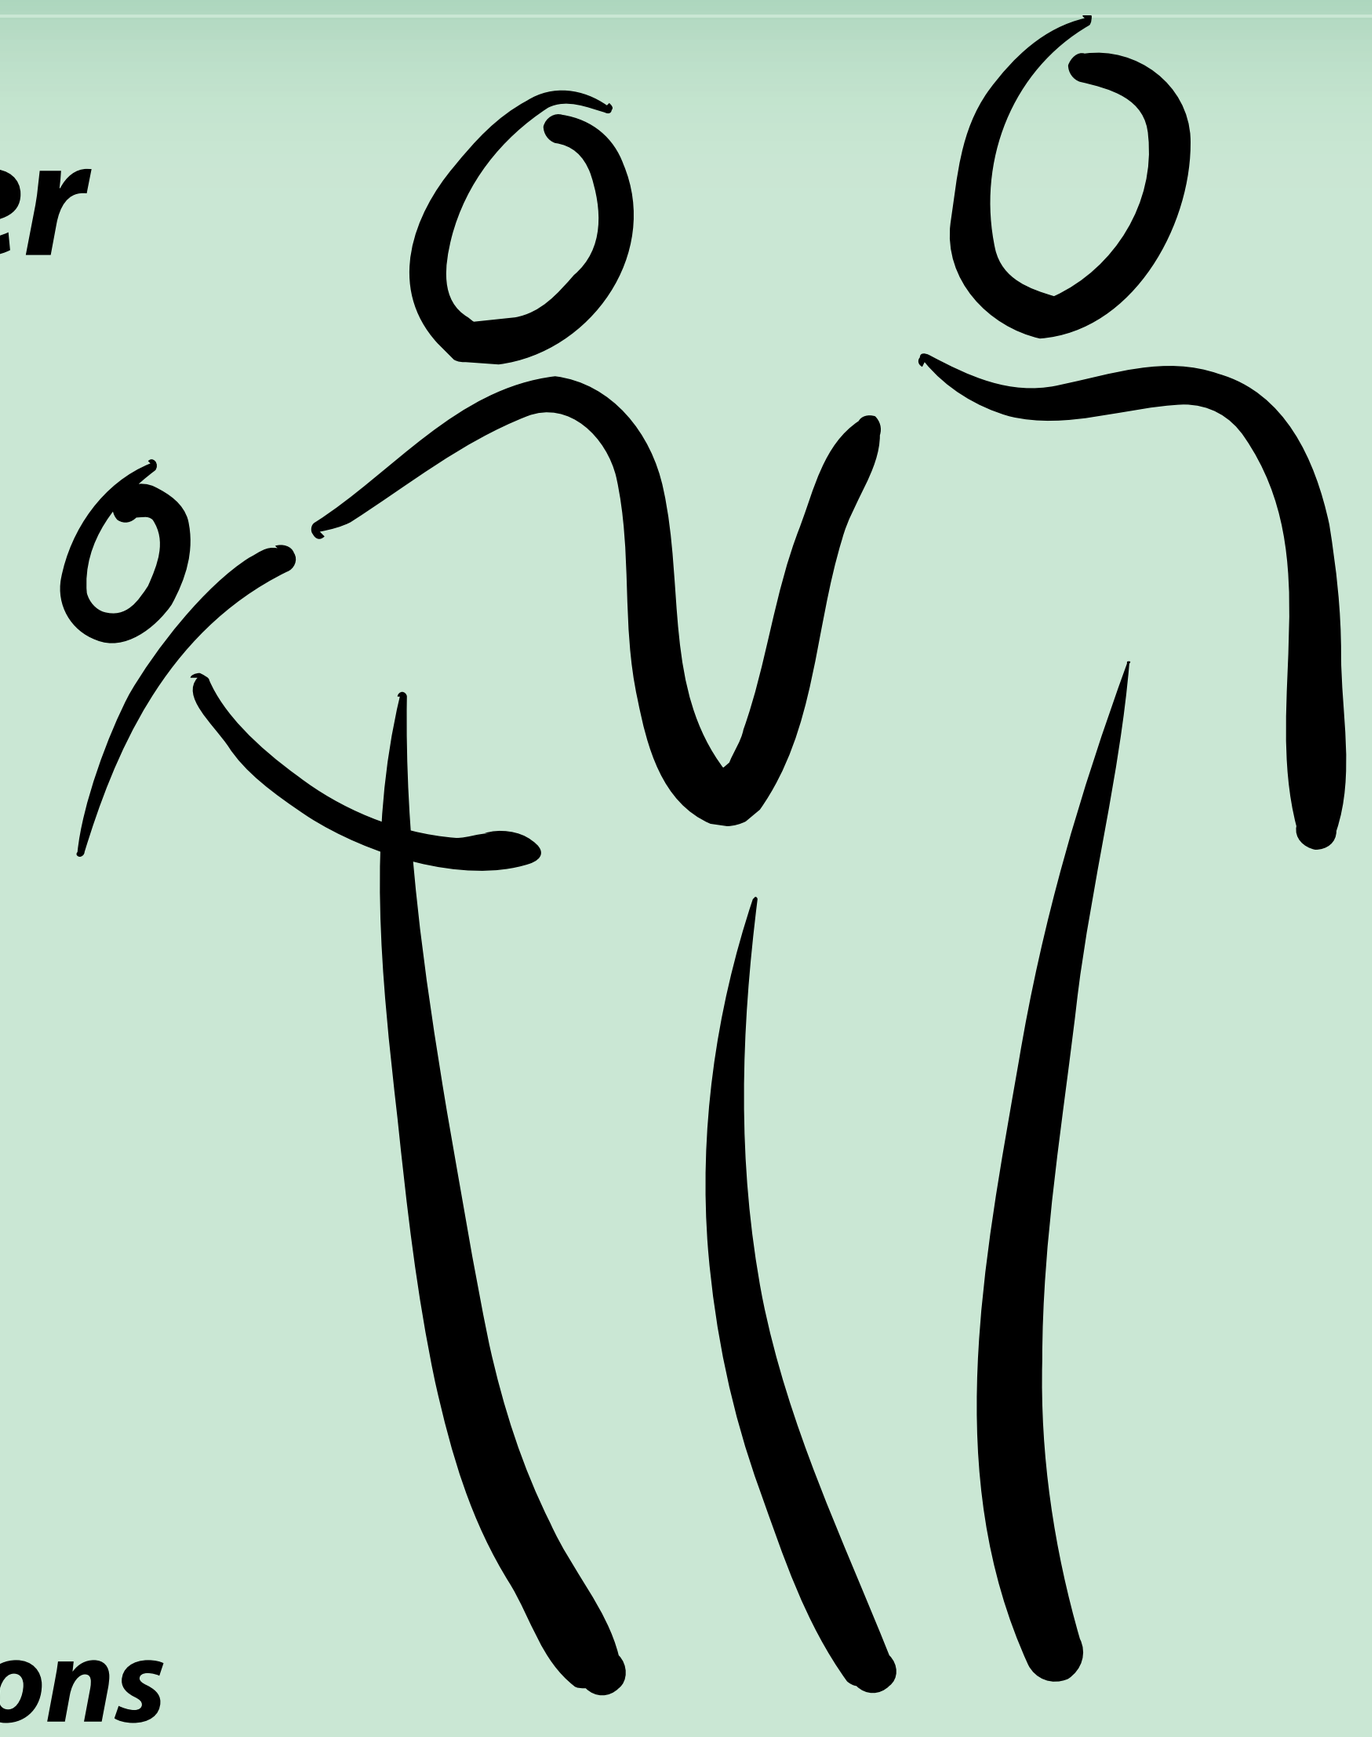

Background

Progesterone production during the luteal phase is necessary for successful implantation of developing embryos. Some components of assisted reproduction, such as GnRHa administration and aspiration of granulosa cells along with mature oocytes, can interfere with the natural production of progesterone. Therefore, supplemental progesterone for luteal phase support is typically administered to patients following in vitro fertilization and embryo transfer (IVF-ET). Luteal phase progesterone supplementation has been shown to result in significantly increased pregnancy rates (Daya and Gunby 2004). However, the relative efficacy of alternative routes of administration is unknown. The objective of this study was to evaluate the effectiveness of vaginal progesterone supplementation compared to intramuscular (IM) progesterone supplementation for luteal phase support after IVF-ET.

Materials and Methods

In a separate study, forty patients were randomized to one of three forms of vaginal progesterone supplementation for luteal phase support after IVF-ET. Endometrin® 100 mg (a novel formulation of progesterone, developed by Ferring Pharmaceuticals, Inc.) was received BID by 12 patients, and TID by 11 patients, and Crinone® 8% gel 90 mg was received QD by 17 patients. Treatment was initiated on the day following oocyte retrieval and continued for 10 weeks.

Pregnancy outcomes were compared to outcomes of IVF-ET cycles conducted concurrently at the same ART center, in which patients were treated with our standard protocol for luteal phase support using IM progesterone. In addition to progesterone, all patients received concomitant estrogen supplementation during the luteal phase with Estrace® 2 mg po BID starting the evening of retrieval. Ovarian stimulation was by FSH/hMG with a LDR/LPL or Lupron® stop protocol for all cycles in both treatment groups. Controls receiving IM progesterone were matched to case cycles for age, baseline FSH, endometrial thickness on the day of hCG trigger injection, stage of embryo transfer (cleavage or blastocyst), and number of embryos transferred. Each case cycle was matched to 5 control cycles, for a total of 200 control cycles. Rates of clinical intrauterine pregnancy (defined by ultrasound identification of a gestational sac), miscarriage, and live birth were compared between case and control cycles by χ^2 analysis.

Results

Clinical outcomes were similar regardless of the type of vaginal progesterone supplementation used (**Table 1**). The different types of vaginal progesterone supplementation were therefore pooled for comparison with patients receiving IM progesterone supplementation.

Table 1. Patient and cycle characteristics and clinical outcomes of treatment cycles compared among three forms of vaginal progesterone supplementation.

	Endometrin® BID	Endometrin® TID	Crinone® QD
Number of cycles	12	11	17
Patient age (years)	35.2 ± 4.5	35.0 ± 5.9	36.0 ± 3.8
Baseline FSH (IU/L)	7.4 ± 1.5	7.1 ± 2.4	7.4 ± 1.8
Endometrium (mm)	11.4 ± 2.2	12.9 ± 3.4	12.0 ± 2.8
Embryos per ET	2.4 ± 0.2	2.3 ± 0.2	1.9 ± 0.1
CIUP per cycle	50.0%	45.4%	52.9%
SAB	1 (@17-20w)	0	0

The matching of patients receiving vaginal progesterone supplementation to control patients receiving the standard IM progesterone was very effective. Patient age, baseline FSH, endometrial thickness, developmental stage at ET, and the number of embryos transferred were all very similar between the two treatment groups (**Table 2**), with no differences approaching statistical significance.

Table 2. Patient and cycle characteristics compared between vaginal and intramuscular progesterone treatment groups.

	Vaginal progesterone	Intramuscular progesterone
Number of cycles	40	200
Patient age (years)	35.5 ± 4.5	35.4 ± 4.4
Baseline FSH (IU/L)	7.3 ± 1.8	7.4 ± 1.7
Endometrium (mm)	12.1 ± 2.8	11.8 ± 2.4
ET stage (% blastocyst)	35.0	35.0
Embryos per ET	2.2 ± 0.5	2.3 ± 0.6

Pregnancy and Live Birth

Outcomes were also very similar between treatment groups. The live birth rate was high (47%) and nearly identical between treatment groups (**Figure 1**). There were 20 pregnancies (50.0%) among patients treated with vaginal progesterone, and 103 pregnancies (51.5%) among matched patients treated with IM progesterone. Among the patients treated with vaginal progesterone, there were no first trimester miscarriages and 1 second trimester miscarriage, while among the patients treated with IM progesterone there were 6 first trimester losses and 3 second trimester losses (**Figure 2**). There were no significant differences in treatment outcomes between groups.

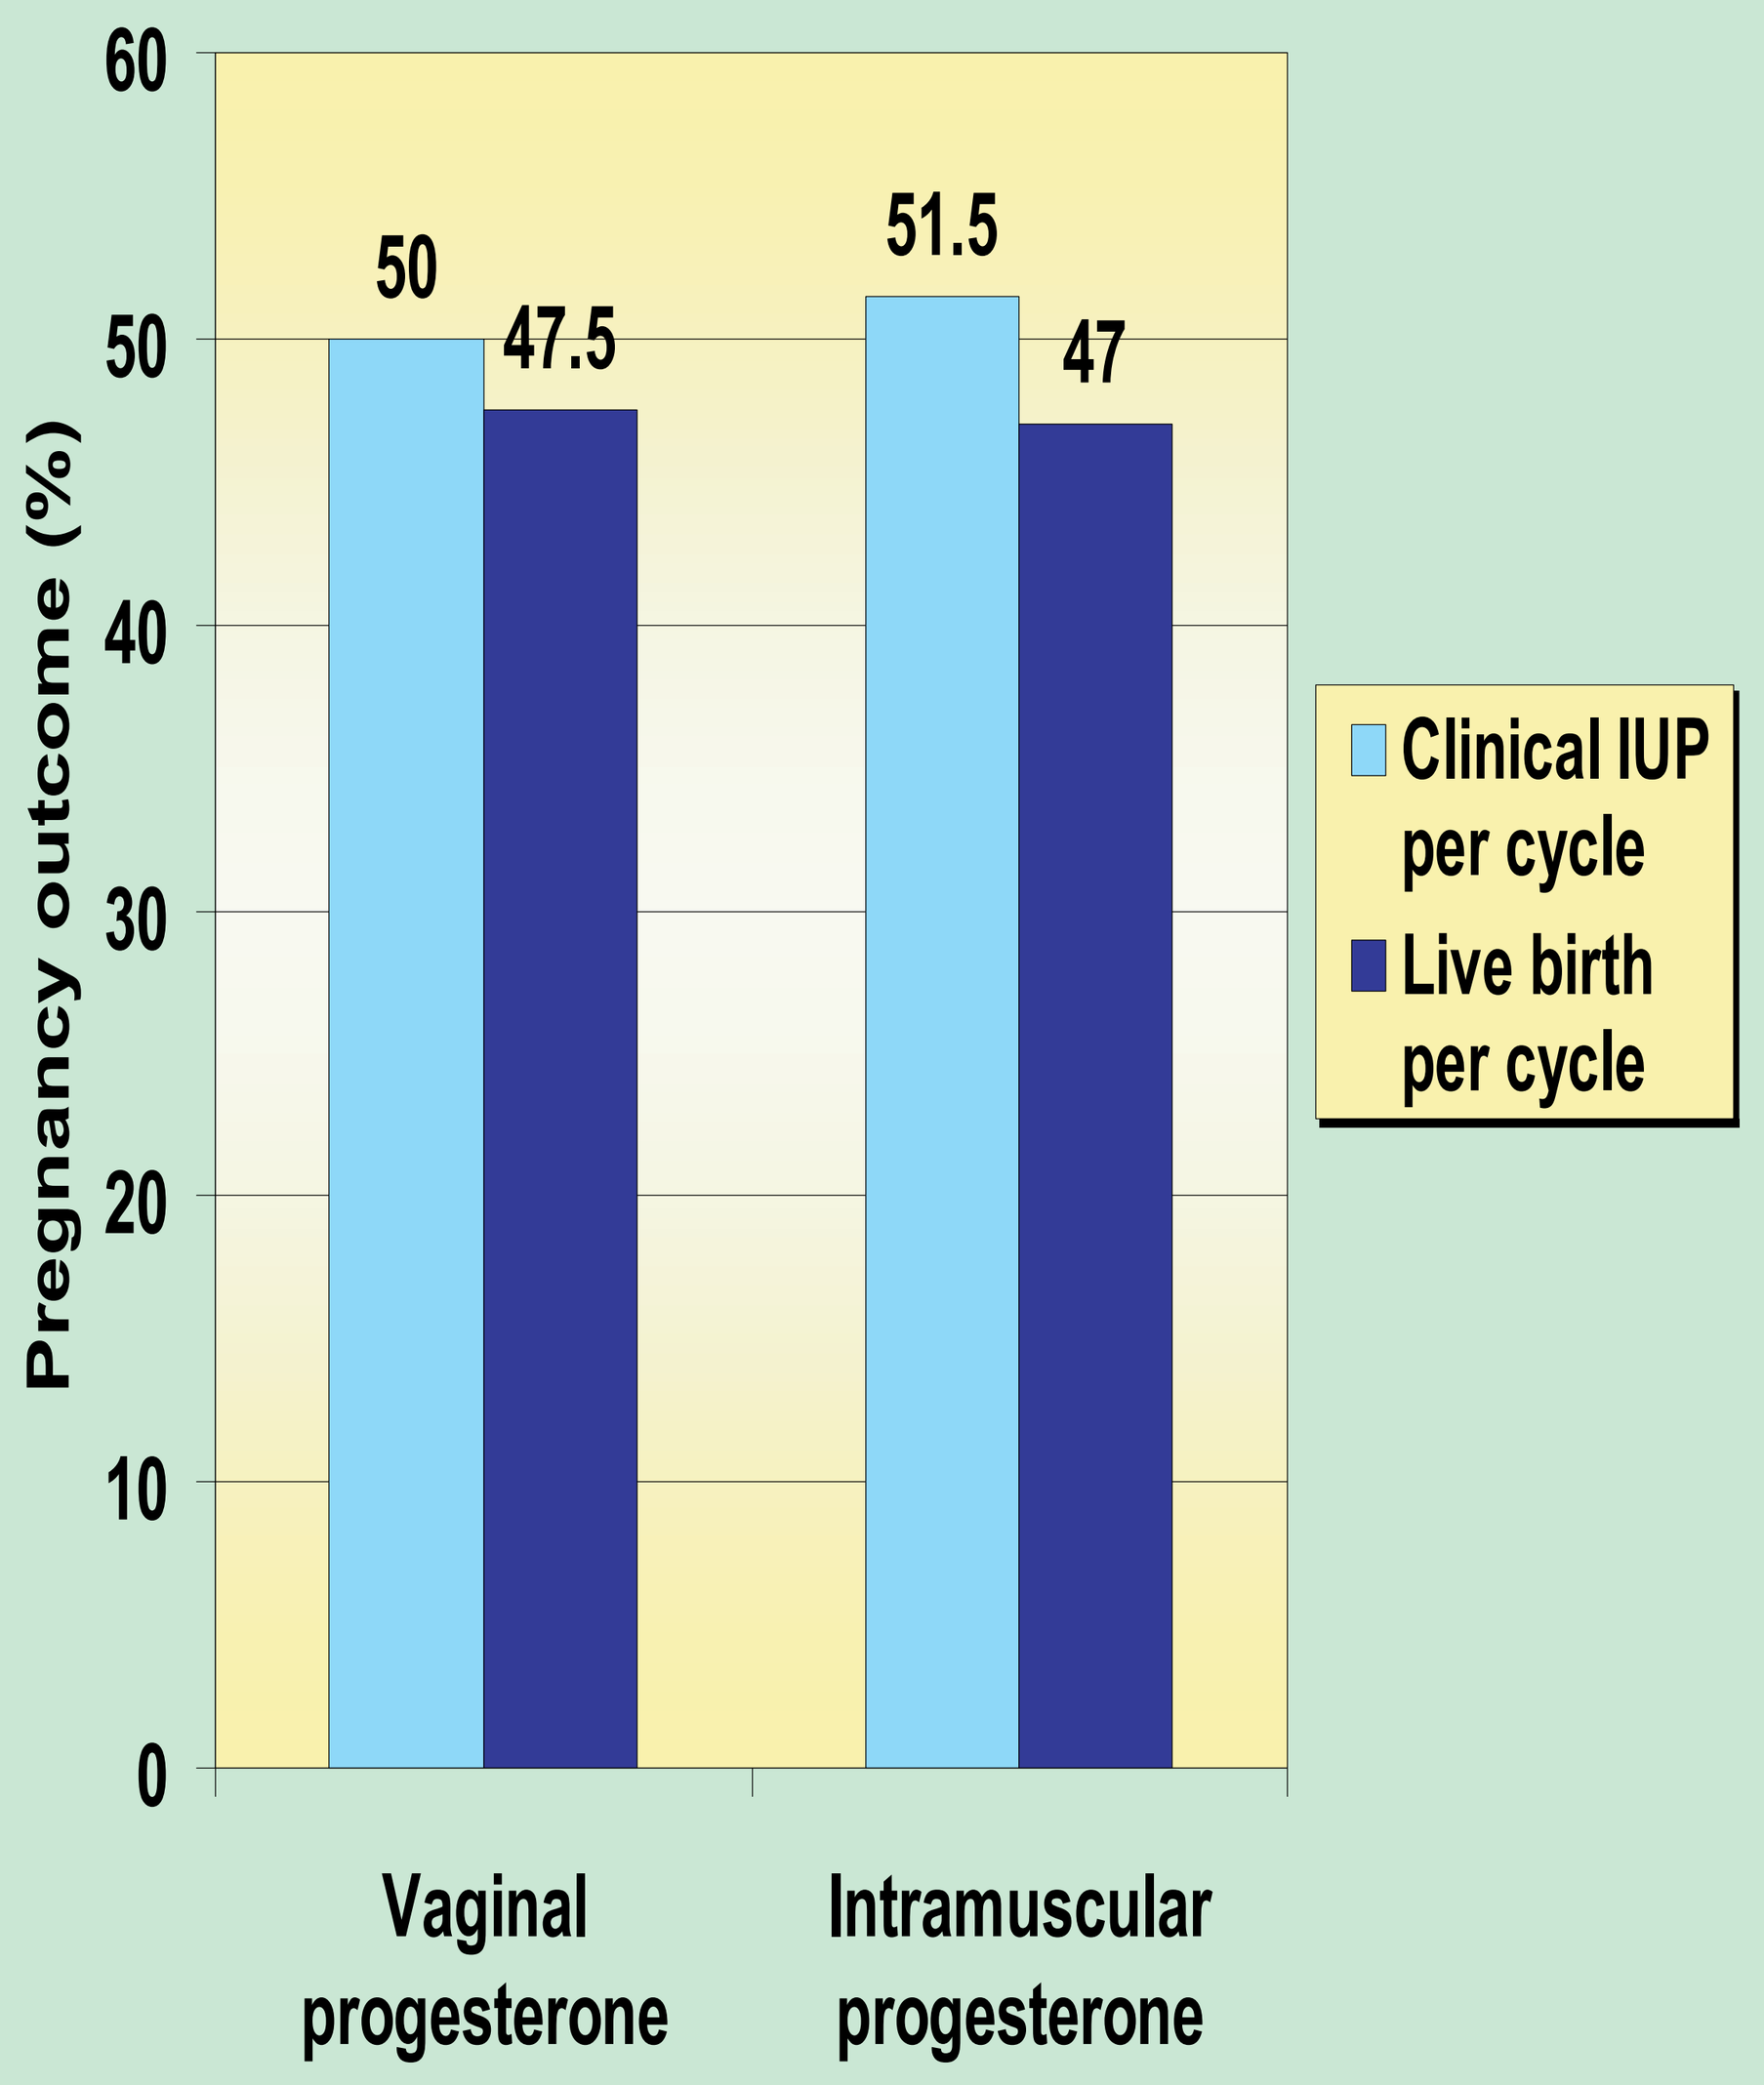
60
50
40
30
20
10
0
Pregnancy outcome (%)
50
47.5
51.5
47
Clinical IUP per cycle
Live birth per cycle
Vaginal progesterone
Intramuscular progesterone

Figure 1. Pregnancy and live birth rates compared between vaginal and IM progesterone treatment groups.

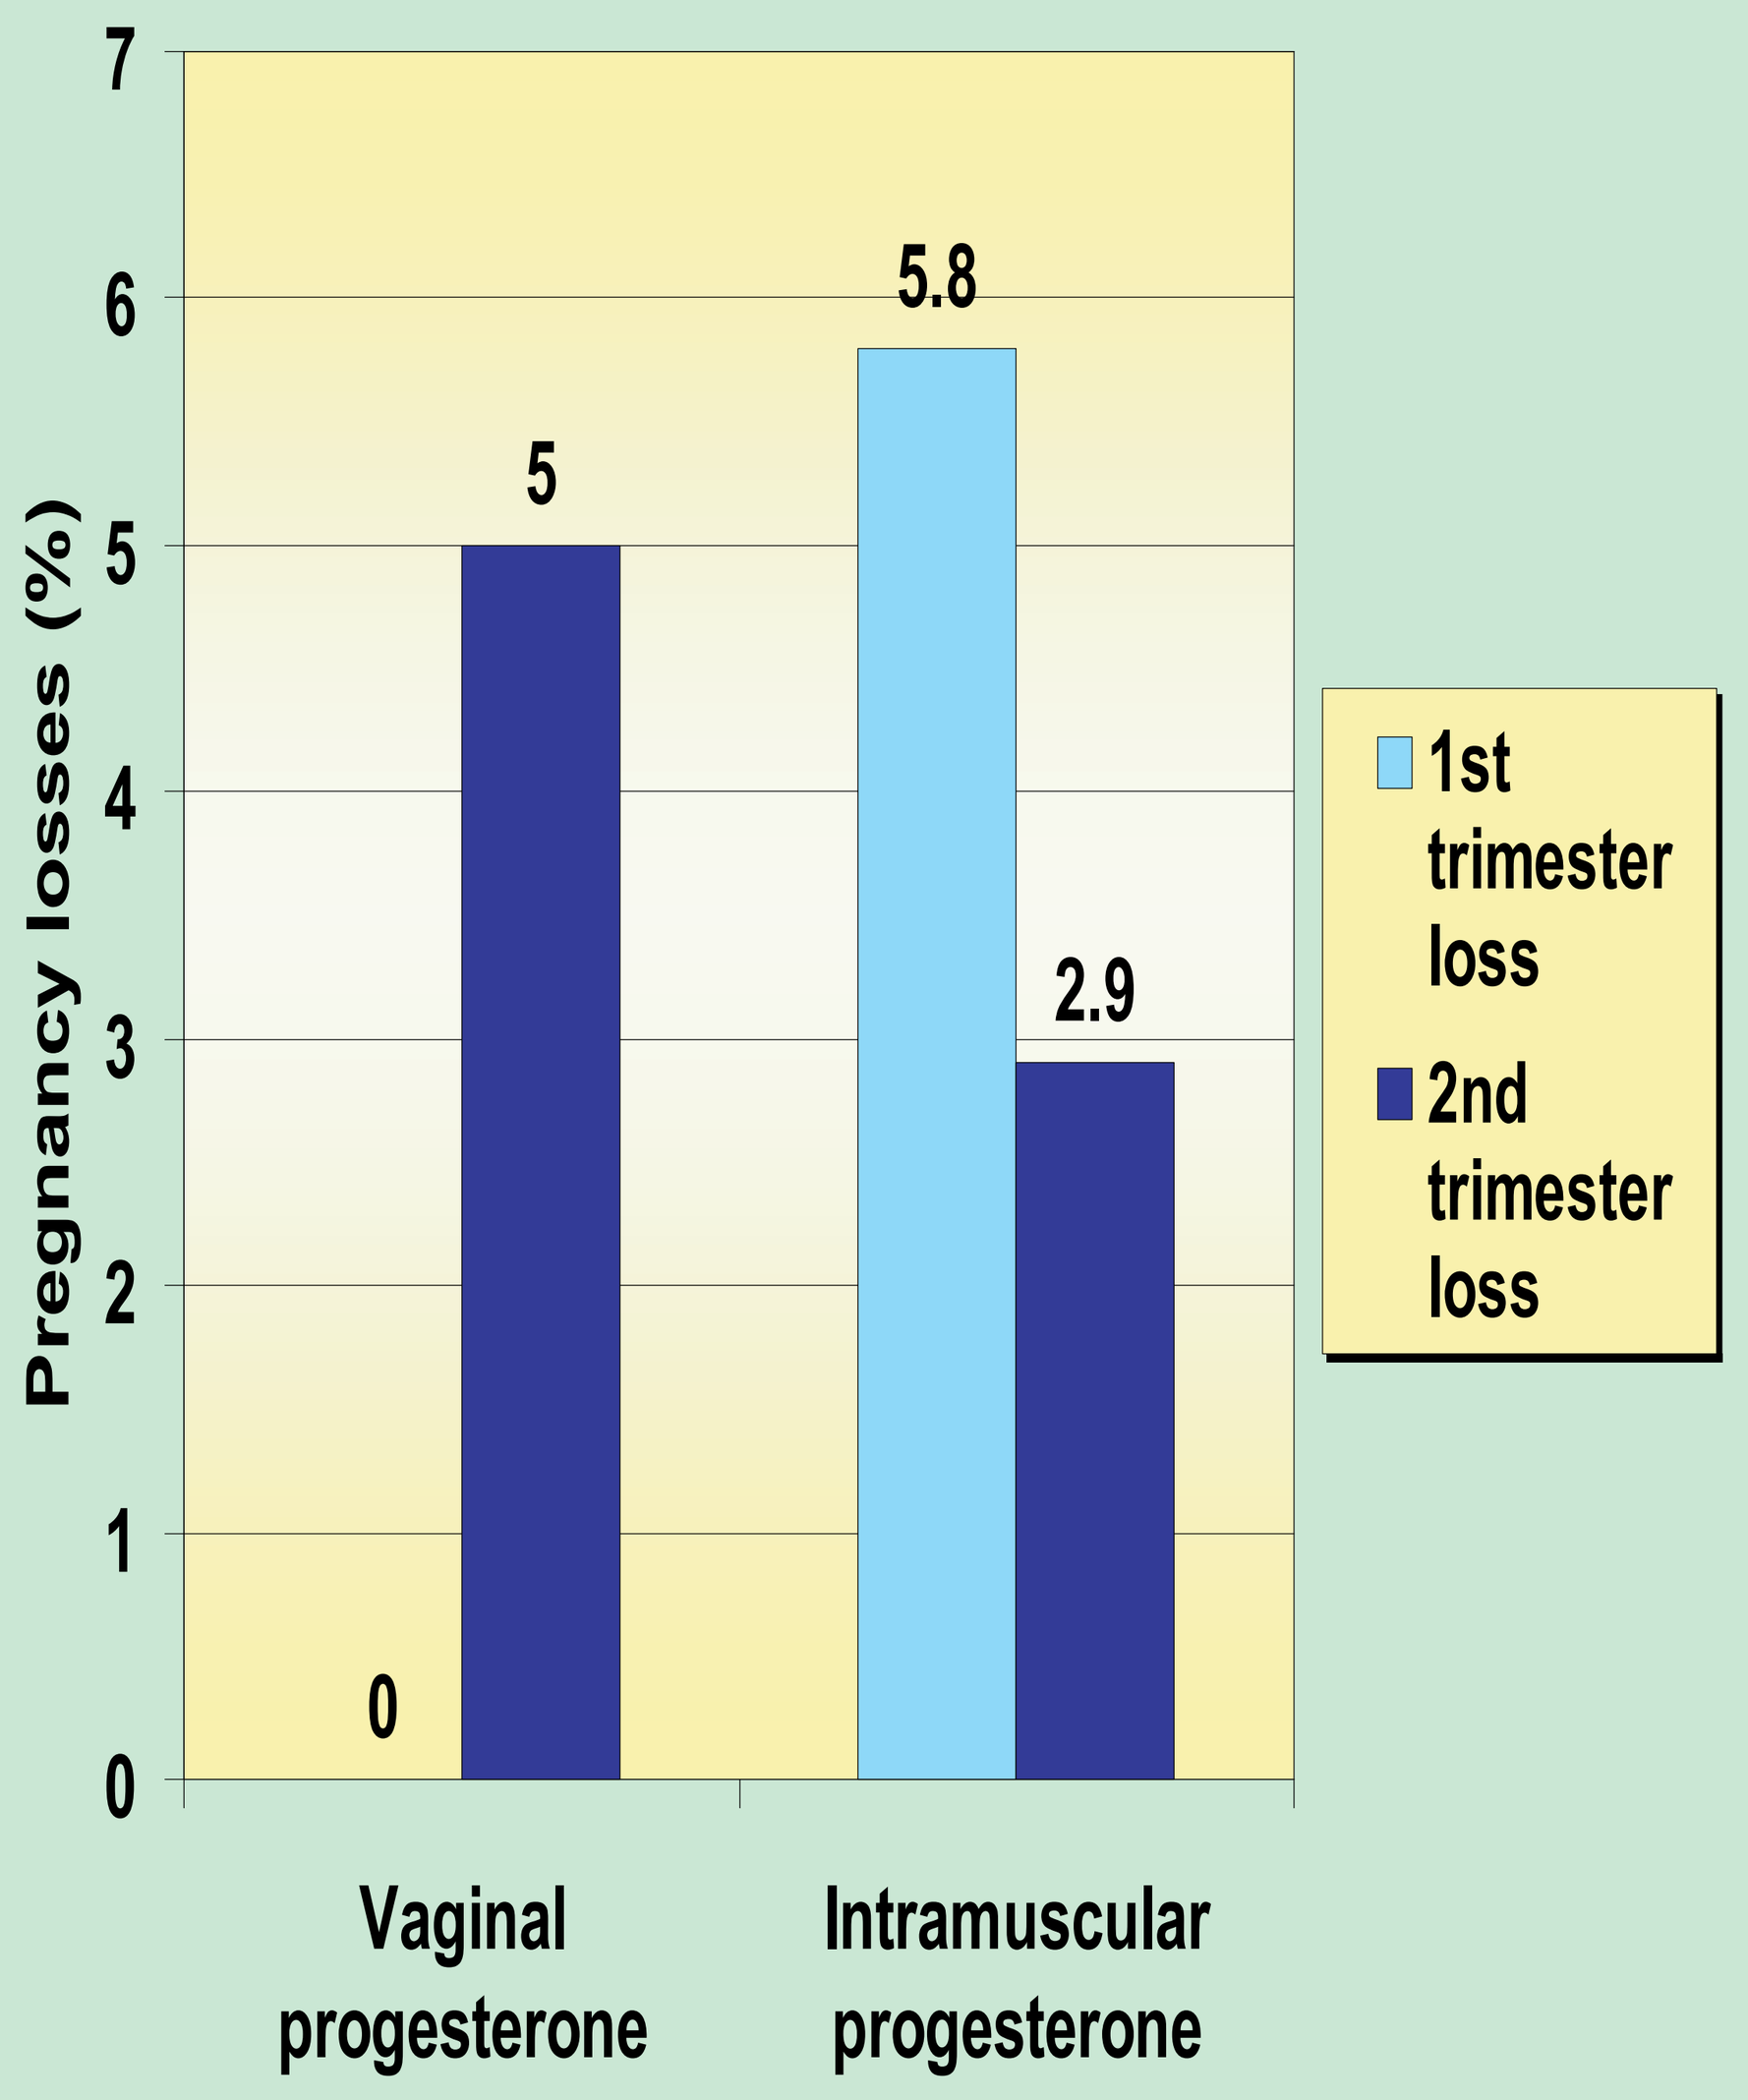
7
6
5
4
3
2
1
0
Pregnancy losses (%)
0
5
5.8
2.9
1st trimester loss
2nd trimester loss
Vaginal progesterone
Intramuscular progesterone

Figure 2. First and second trimester pregnancy losses compared between vaginal and IM progesterone treatment groups.

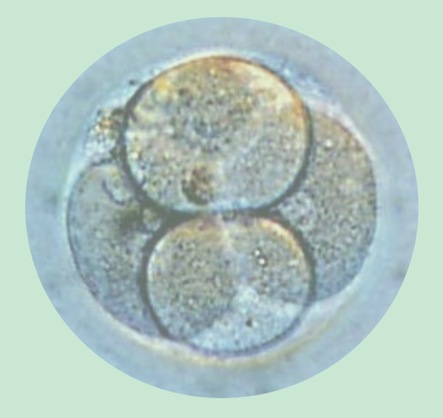
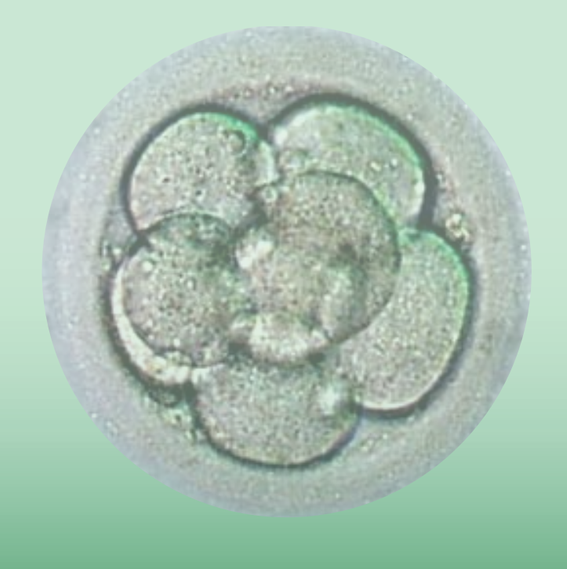
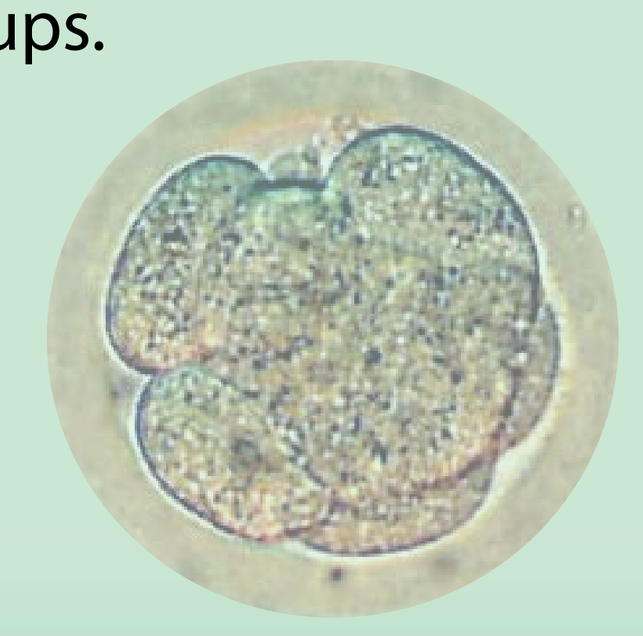
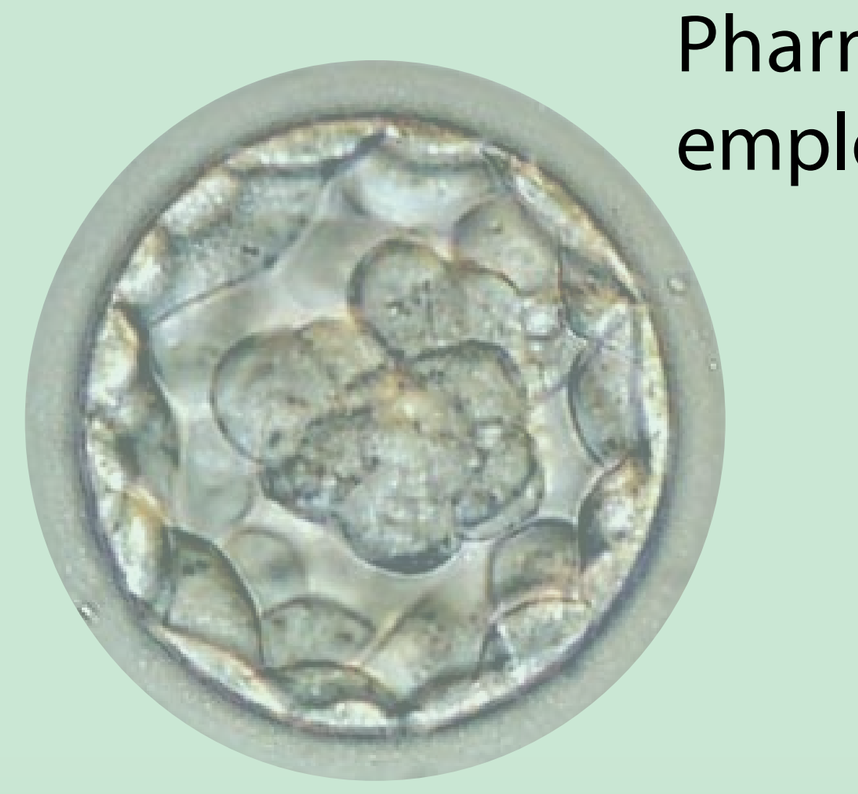
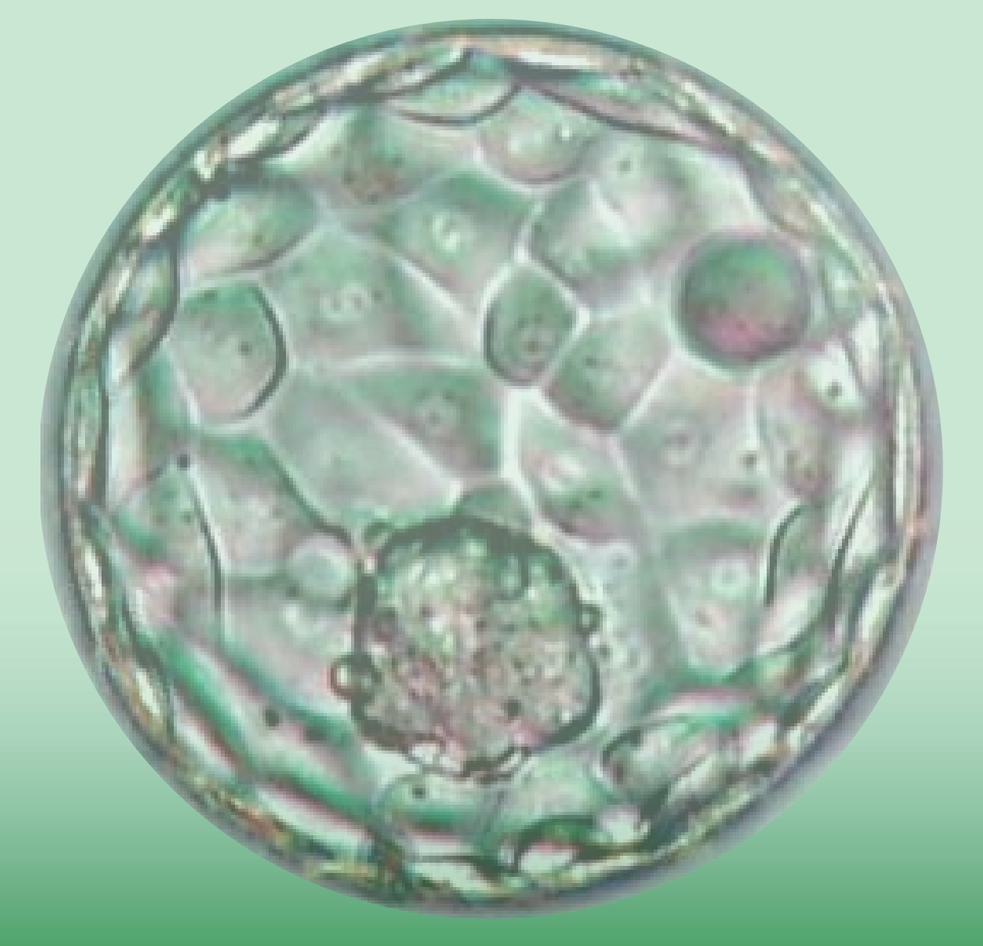

Conclusions

Results of this study suggest that rates of pregnancy, miscarriage, and live birth are similar between patients treated with vaginal progesterone and those treated with IM progesterone for luteal phase support after IVF-ET. However, larger scale randomized studies are needed to confirm the equivalence of these two treatments methodologies. In the absence of clear evidence demonstrating a clinically significant difference in efficacy, the choice of the route of progesterone administration may be based on other criteria such as tolerability.

Reference

Daya S, Gunby J. Luteal phase support in assisted reproduction cycles. Cochrane Database of Systematic Reviews 2004, Issue 3. Art. No.: CD004830

Disclosures

Endometrin® is not currently approved for use by the FDA. Luteal phase support is an off-label use of IM progesterone. The randomized study of different forms of vaginal progesterone supplementation was sponsored by Ferring Pharmaceuticals, Inc. Drs. Blake and Yankov are employees of Ferring Pharmaceuticals, Inc.