Endometrin® for luteal phase support in a randomized, controlled, open-label, prospective IVF clinical trial using a combination of Menopur® and Bravelle®: comparison by age, BMI and basal FSH

William Schoolcraft, MD, Colorado Center for Reproductive Medicine, Englewood, CO; Charles Miller, MD, Charles Miller & Associates, Arlington Heights, IL; Jerome H. Check, MD, PhD, Cooper Institute for Reproductive Hormonal Disorders, Marlton, NJ; Russell Foulk, MD, Nevada Center for Reproductive Medicine, Reno, NV; Bradford Kolb, MD, Huntington Reproductive Center, Pasadena, CA; Anthony N. Wakim, MD, Magee Women's Hospital, Pittsburgh, PA

INTRODUCTION

In vitro fertilization (IVF) and other assisted reproductive technology (ART) treatments often involve suppression of normal ovarian function and should take into consideration idiopathic endocrine function. Progesterone supplementation during the luteal phase of IVF cycles has become a standard procedure (1, 2) to supplement endogenous contributions from the corpus luteum that may have been compromised during ovulation induction or oocyte retrieval.

Exogenous progesterone can be administered orally, vaginally, and by intramuscular injection. In order to circumvent the first-pass liver effect, oral administration requires high doses which can result in progestational side effects. Intramuscular (IM) injections are not FDA-approved and are painful. Vaginal formulations are more comfortable and convenient to administer and, in addition, may permit targeted drug delivery to the uterus (3), since it has been theorized that absorption through the vaginal walls could deliver high levels of medication directly to the uterine circulation, thereby providing high levels of progesterone to the endometrial tissue.

The objective of this study was to compare the safety and efficacy of a micronized, naturally-derived vaginal progesterone tablet insert, Endometrin® 100 mg (Ferring Pharmaceuticals, Inc., Suffern, NY, USA), with a vaginal progesterone gel, Crinone® 8% 90 mg (Columbia Laboratories, Inc., Livingston, NJ, USA) in women undergoing IVF.

MATERIALS AND METHODS

This multicenter, randomized, open-label (assessor-blinded), parallel-group Phase III study in women undergoing IVF was conducted to compare the pregnancy rates following luteal phase support with micronized, naturally-derived vaginal progesterone tablet inserts (Endometrin® 100 mg BID or TID) or vaginal progesterone gel (Crinone® 8% 90 mg QD). The study was conducted at 25 sites in the United States between July 2005 and April 2006.

Each study center followed the IVF protocol guidelines in practice at that site. Patients who conceived continued on study drug for approximately 10 weeks.

PATIENT SELECTION

Healthy, premenopausal women between 18 and 42 years of age, with a BMI ≤34 kg/m^2, a baseline FSH ≤15 IU/L, a history of infertility, and requiring IVF, were eligible to enroll in the study.

STUDY PROCEDURES

Pituitary down-regulation was performed with an injectable gonadotropin-releasing hormone (GnRH) agonist using a long protocol with luteal phase initiation. Documentation of down-regulation, indicated by a serum estradiol level of <50 pg/mL, endometrial lining of <7 mm, and no evidence of ovary activity on transvaginal ultrasound (TVU), was required prior to the start of gonadotropin treatment. Gonadotropin therapy was done in accordance with the usual IVF protocol at each center, with the only requirement being that a minimum of one vial of hMG (Menopur®) had to be used daily. All centers chose to use Menopur® and Bravelle® from Day 1. In addition, the centers had the option of combining the FSH and hMG and administering them as one single daily injection. This was done for approximately two-thirds of the patients (n=777). Gonadotropin stimulation was stopped when the lead follicle mean diameter was at least 18 mm, and a single intramuscular injection of human chorionic gonadotropin (hCG) (Novarel®) was administered to trigger ovulation. Oocytes were retrieved within approximately 36 hours after Novarel® administration according to site-specific procedures. The only requirement for embryo transfer was that no more than three embryos could be transferred if done on Day 3 post-retrieval and no more than two blastocysts could be transferred if done on Day 5 post-retrieval.

Patients were randomly assigned to one of three treatment regimens: Endometrin® 100 mg BID, Endometrin® 100 mg TID, or vaginal progesterone gel 90 mg QD. Randomization occurred the day of or day after oocyte retrieval, with study drug (Endometrin® BID, Endometrin® TID, or vaginal gel QD) initiated on the day after oocyte retrieval. Given the impact of age and ovarian reserve on response to hormonal treatment, the study was randomized and stratified *a priori* for age (<35, 35-37, 38-40, and 41-42 years) and ovarian reserve as reflected in baseline FSH level (<10 and 10-15 IU/L). Embryo transfer generally occurred on Day 3 or Day 5, according to the site's IVF guidelines.

Although study drug was administered open-label, it was assessor-blinded; the person who performed the TVUs to confirm clinical and ongoing pregnancy was blinded to the patient's treatment group assignment. The patients who were pregnant continued on the study medication for ten weeks. A final visit was scheduled upon completion of the tenth week of study drug (on the day of the last dose or within three days from the last dose).

OUTCOME MEASURES

The efficacy variables of pregnancy and live birth rates were determined.

STATISTICAL ANALYSIS

The primary analysis was performed to determine if the ongoing pregnancy rate for each Endometrin® regimen was non-inferior to vaginal gel, using a two-sided 95% confidence interval for the difference in pregnancy rates and an arbitrary prespecified non-inferiority lower bound of 10%.

RESULTS

A total of 1211 patients underwent oocyte retrieval and were randomized to treatment; 404 patients each were randomized to Endometrin® BID and Endometrin® TID, and 403 patients were randomized to vaginal progesterone gel QD. Patients ranged in age from 19 to 42 years. Only data for those patients who received embryo transfers (the efficacy population) are presented, in order to focus on any differences among the different luteal support regimens.

All patients were treated with a combination of Bravelle® and Menopur® during the stimulation phase. Patients responded well to this gonadotropin combination and the results can be seen in Table 1. Patients were stimulated for an average of 10.2 days, with a mean of 13.8 oocytes retrieved.

TABLE 1. OVARIAN STIMULATION OUTCOMES

PARAMETER	ALL PATIENTS
	(n=1,211)
Duration of treatment (days)	10.2
Mean number of oocytes retrieved	13.8
Patients undergoing embryo transfer	97%
Number of embryos transferred	2.4
Day 3	2.6
Day 5	2.0

The results broken out by age, FSH and BMI can be seen in Table 2 and Figures 1 and 2. The well-established influence of age on pregnancy outcomes (4) was evident in the current data, in that patients >35 years of age did not achieve a pregnancy as often as the younger group; however, the pregnancy rates were still excellent in this group and higher than SART data (4) **(Table 2)**. Baseline FSH values did not significantly impact pregnancy outcomes, as there was not a significant drop in pregnancy rates in patients with elevated basal FSH (>10 IU/L) compared to normal FSH values **(Figure 1)**. The results of the patients segregated by BMI demonstrated no discernible difference between patients with high BMI (>30 kg/m^2) versus patients within the normal range **(Figure 2)**. While the results were highly variable in the group with BMI <18.5, it is worth noting that this represented a very small number of patients: six patients in the Endometrin BID group, and ten patients in each of the Endometrin TID group and the vaginal gel group.

TABLE 2. CONTINUING PREGNANCY RATES BY AGE

AGE	Endometrin BID N=392	Endometrin TID N=390	Vaginal Gel N=393
<35	111/240	117/236	108/236
35-37	27/87	37/91	41/97
38-40	16/53	12/46	16/53
41-42	2/12	5/17	5/7

FIGURE 1. CONTINUING PREGNANCY OUTCOMES BASED ON BASAL FSH

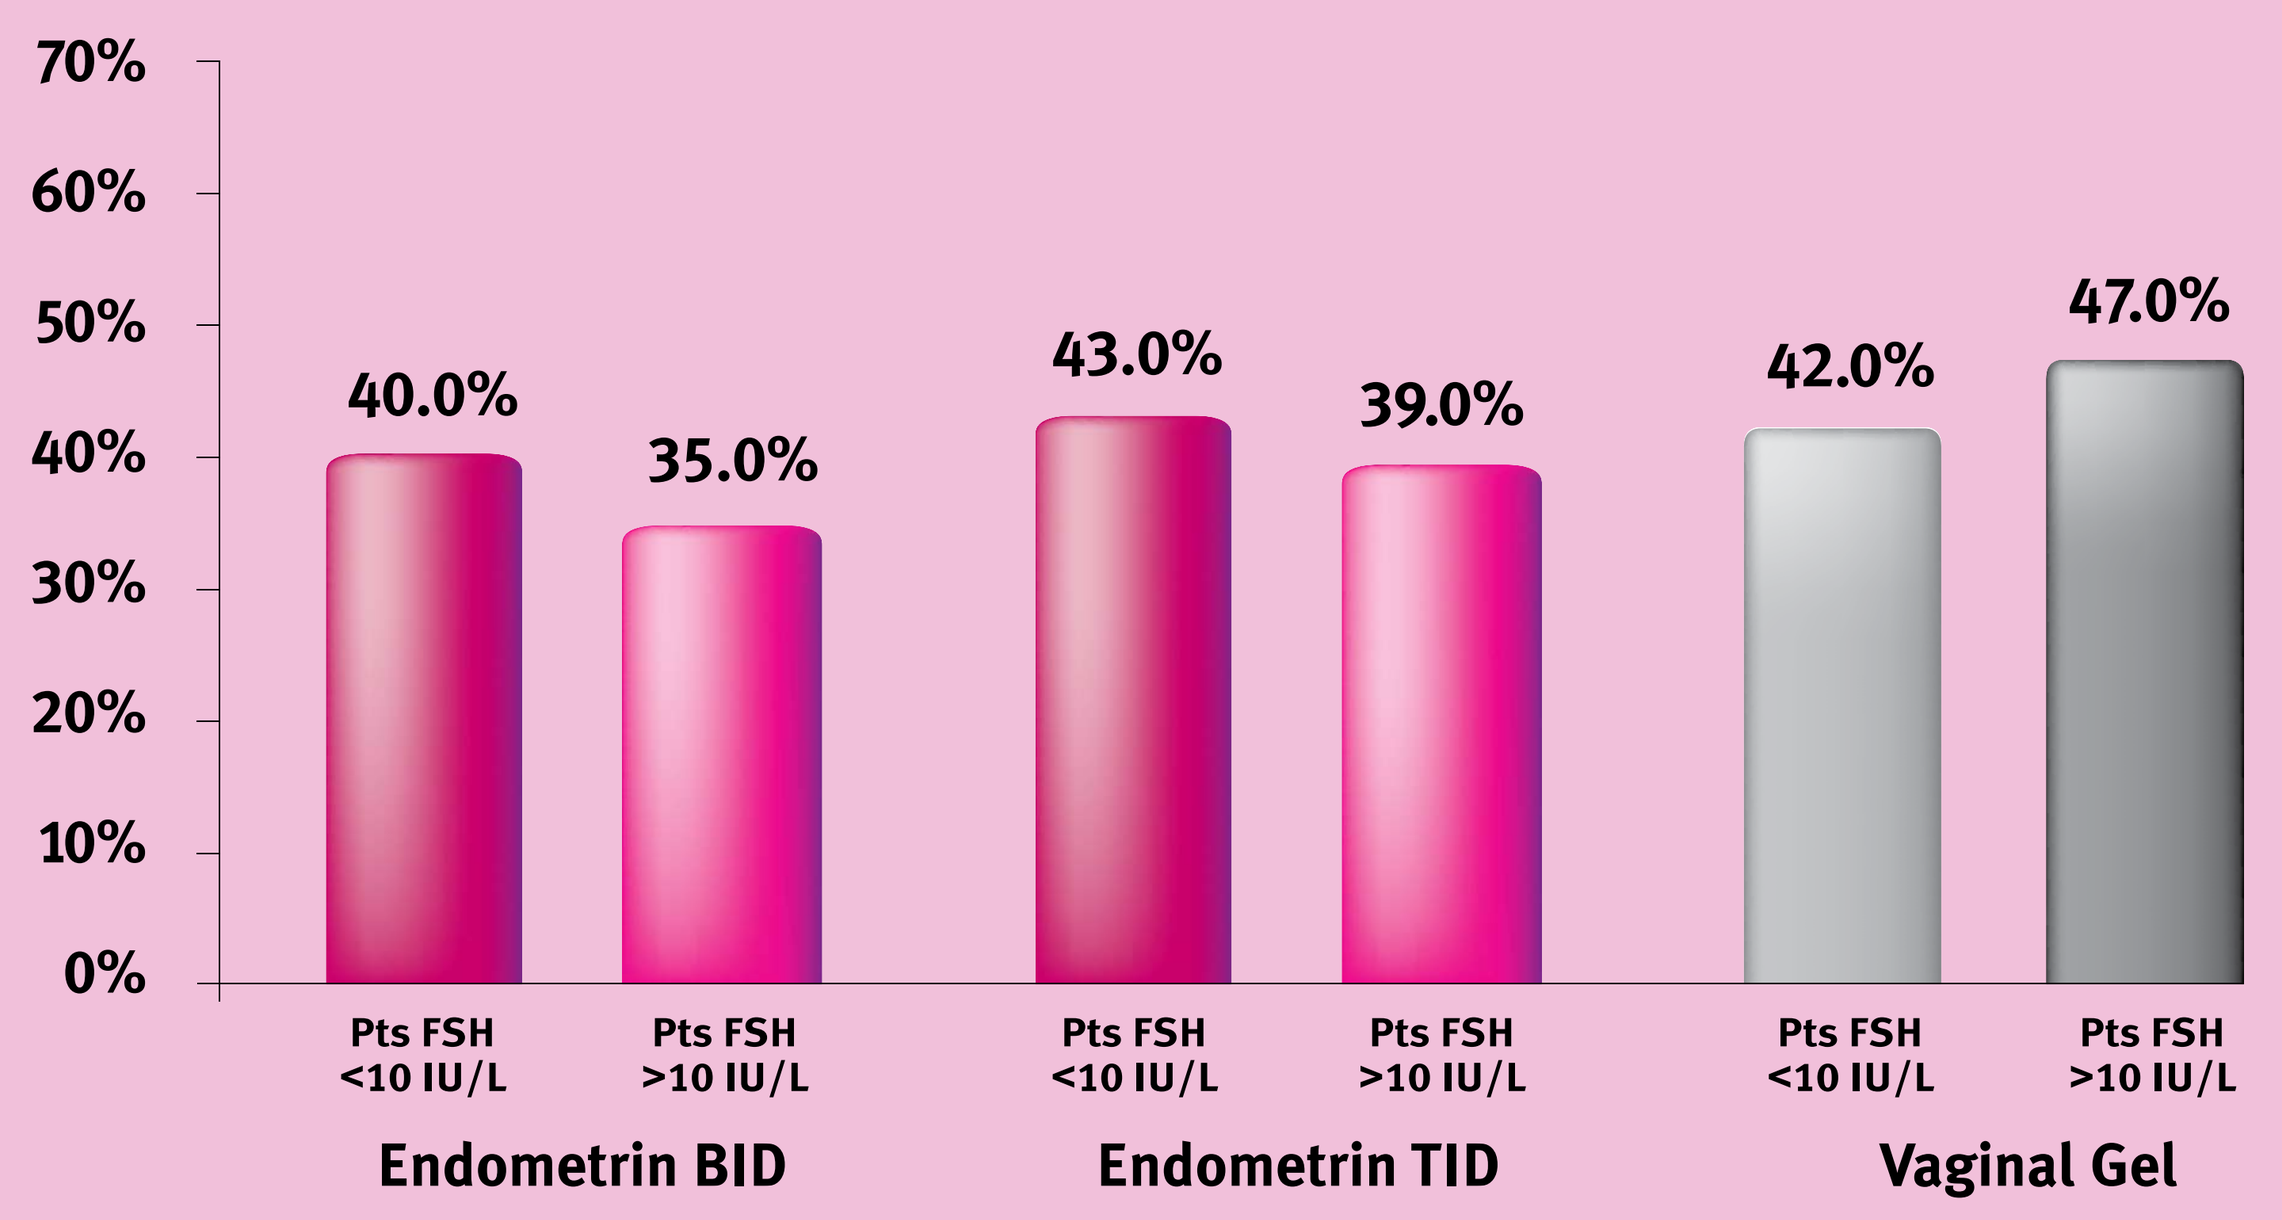
70%
60%
50%
40%
30%
20%
10%
0%
40.0%
35.0%
43.0%
39.0%
42.0%
47.0%
Pts FSH <10 IU/L
Pts FSH >10 IU/L
Endometrin BID
Endometrin TID
Vaginal Gel

FIGURE 2. CONTINUING PREGNANCY OUTCOMES BASED ON BMI

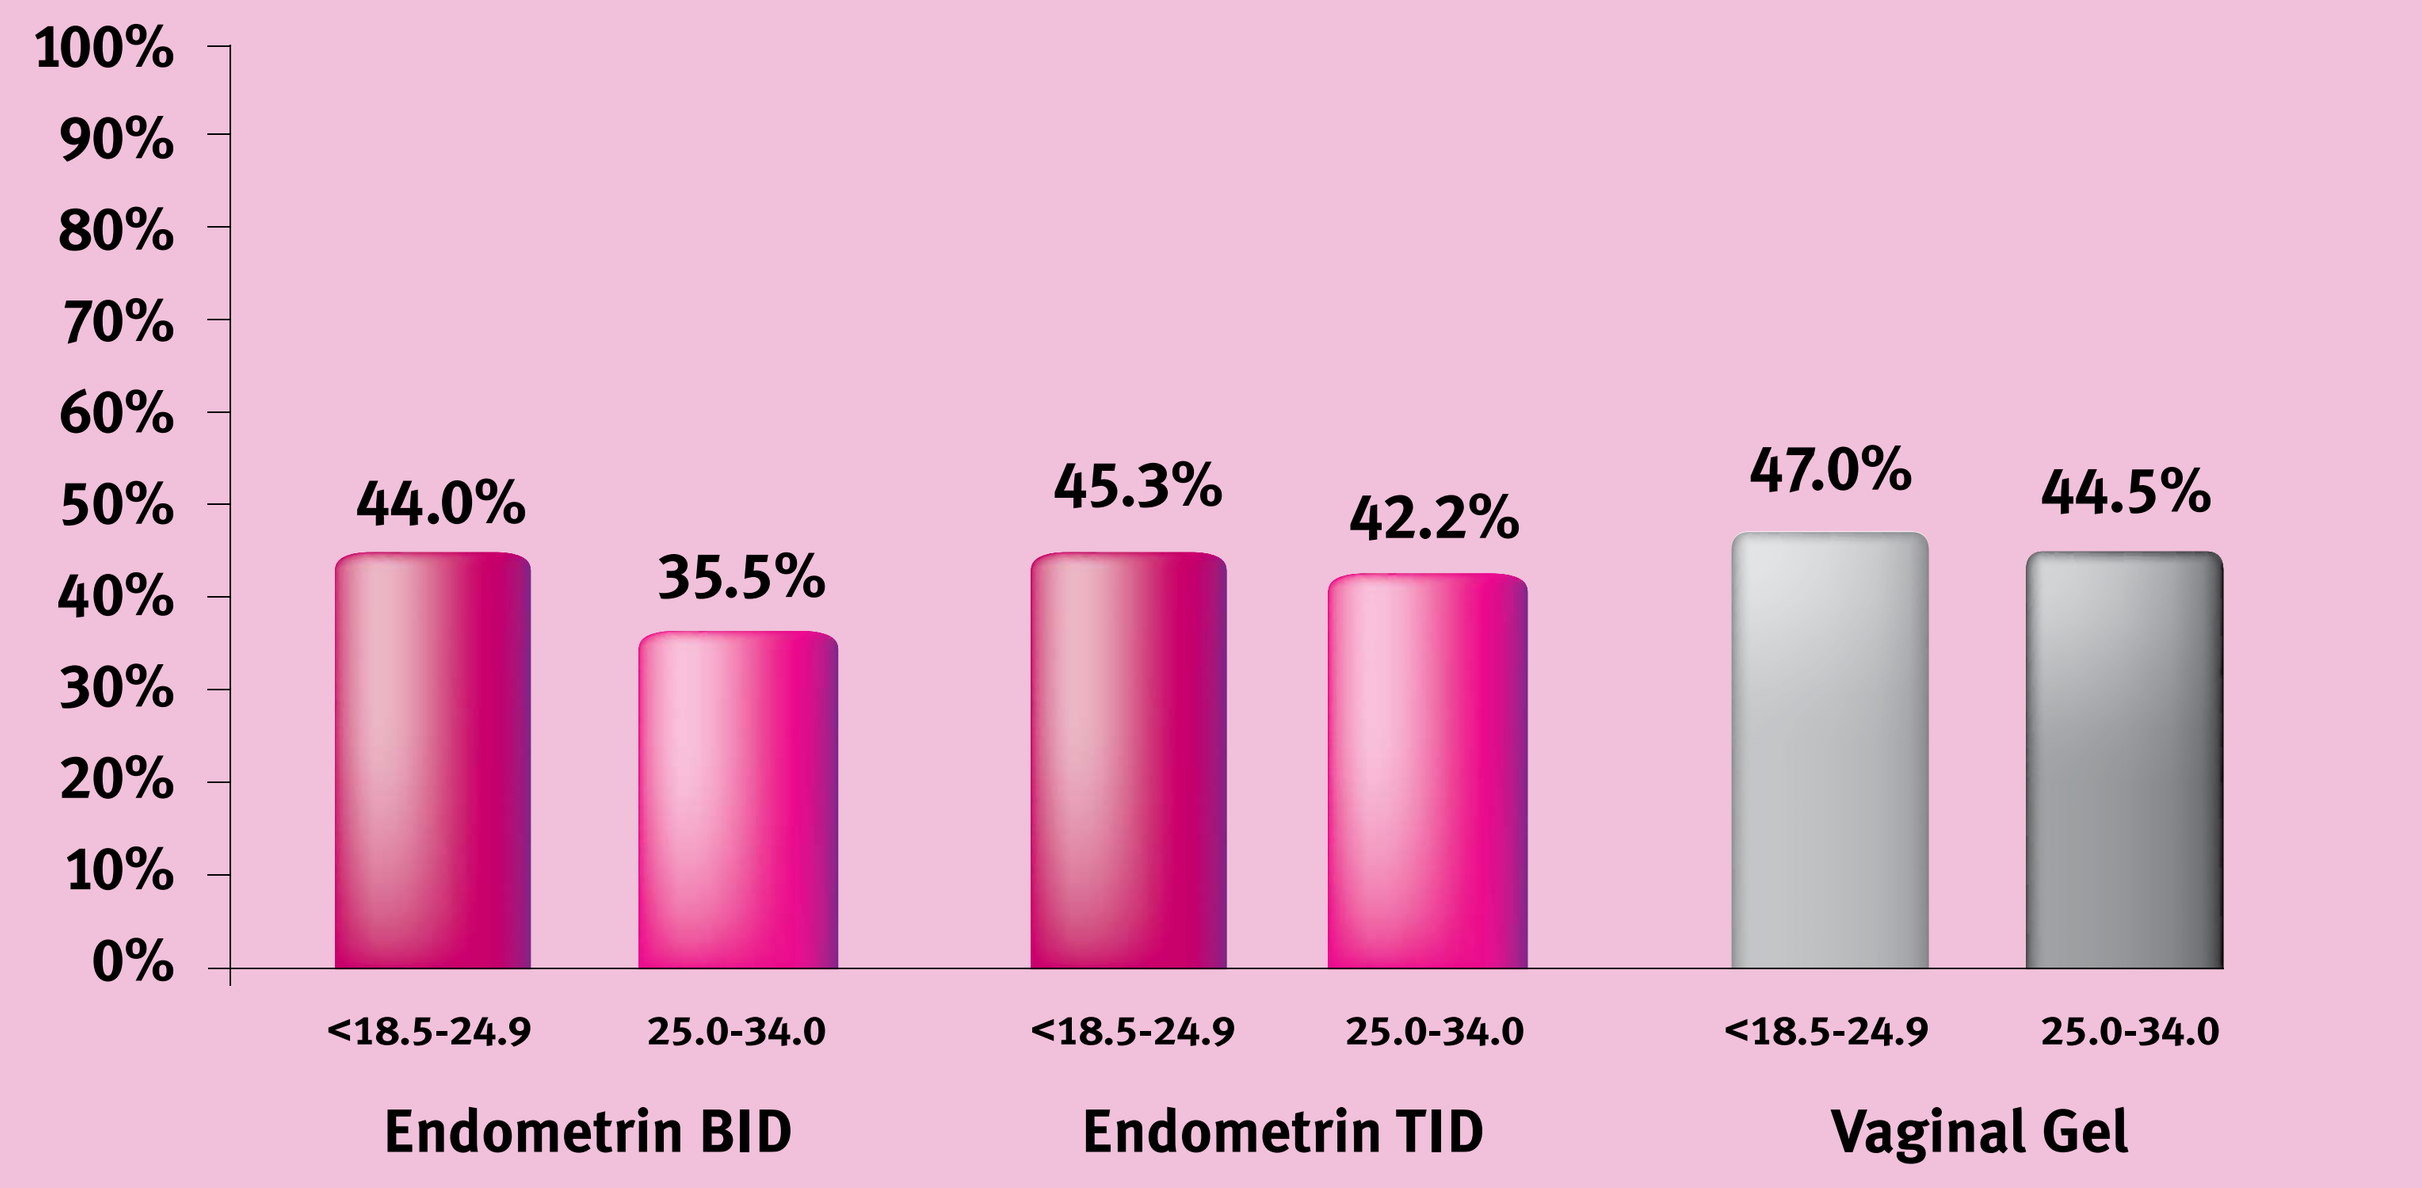
100%
90%
80%
70%
60%
50%
40%
30%
20%
10%
0%
44.0%
35.5%
45.3%
42.2%
47.0%
44.5%
<18.5-24.9
25.0-34.0
Endometrin BID
Endometrin TID
Vaginal Gel

DISCUSSION

Patients undergoing IVF typically receive progesterone supplementation during the luteal phase, a procedure that has been shown to improve pregnancy rates (5). In the current study, patients received progesterone supplementation with either Endometrin®, a micronized, naturally-derived vaginal progesterone tablet insert, or vaginal progesterone gel. Endometrin® was administered either BID or TID and the gel was administered QD per the instructions in the package insert. No clinically meaningful differences were observed across the three treatment groups in biochemical pregnancy rates, clinical pregnancy rates, ongoing pregnancy rates, or live birth rates.

The patients responded well to ovarian stimulation by Bravelle® and Menopur®, which resulted in 97% of patients undergoing embryo transfer.

While there was a drop in pregnancy rates in patients >35 years, the reductions were not as drastic between BMI values, and between basal FSH values. These patients had good pregnancy rates, indicating the treatment paradigm used in this study was efficacious in typically hard-to-treat patients. This effect may also reflect that the participating physicians routinely see these types of patients and have learned how to adjust stimulation protocols to increase success rates.

These data demonstrate that the use of Endometrin® for luteal support in ART patients results in excellent single cycle pregnancy rates and provides more than adequate luteal support based on the live birth rates compared to clinical pregnancy rates. Therefore, Endometrin® provides a safe, well-tolerated, and effective method for providing luteal phase support in women undergoing IVF.

REFERENCES

1. Soliman S, Daya S, Collins J, Hughes EG. The role of luteal phase support in infertility treatment: a meta-analysis of randomized trials. Fertil Steril 1994;61(6):1068-76. **2.** Penzias AS. Luteal phase support. Fertil Steril 2002;77(2):318-23. **3.** Bulletti C, de Ziegler D, Flamigni C, et al. Targeted drug delivery in gynaecology: the first uterine pass effect. Hum Reprod 1997;12(5):1073-9. **4.** Centers for Disease Control and Prevention. 2004 Assisted Reproductive Technology Success Rates. http://ftp.cdc.gov/pub/Publications/art/2004ART508.pdf (Updates posted on http://www.cdc.gov/ART/ART2004) **5.** Pritts EA, Atwood AK. Luteal phase support in infertility treatment: a meta-analysis of the randomized trials. Hum Reprod 2002;17(9):2287-99.